VecTabs® and advanced human *in-vitro* models:
Targeting TDP-43 aggregates and oxidized phosphocholines in Amyotrophic lateral sclerosis

A G Duarte[1,#], S Pasteuning[1], M Spits[1], W Pos[1], F Sharifi[1], S Pascoal[1], R van Dijk[1], M Böhne[1], M Sogorb-Gonzalez[1], B Sanders[1], S van Deventer[1], P S Konstantinova[1]

[1] VectorY BV, Science Park 408, Matrix Innovation Center VI, 1098 XH Amsterdam, The Netherlands

[#] andreia.duarte@vectorytx.com

Introduction

Amyotrophic lateral sclerosis (ALS) is a neurodegenerative disease characterized by the progressive loss of motor neurons, eventually leading to paralysis and premature death. **Nuclear TAR DNA binding protein 43 (TDP-43)** is an RNA binding protein essential for neuronal health, but misfolded and aggregated TDP-43 mis-localizes to the cytoplasm leading to a loss of function and gain of toxicity pathology thought to be involved in **97% of ALS cases**. Misfolded and aggregated TDP-43 is linked to **mitochondrial dysfunction, axonal transport defects**, and to the **neuromuscular junction (NMJ) pathology** that characterizes ALS **(Fig. 1)**. In addition to TDP-43 misfolding, **the main metabolic abnormality in ALS motor neurons** is a dramatically increased production of glycerophospholipids leading to **accumulation of neurotoxic oxidized phosphocholines (OxPCs)** that cause **neuronal death (Fig. 1)**.

We have **developed two therapeutic strategies** for ALS: **(1) AAV-delivered scFv's** (Vectorized Transformative Antibodies, **VecTabs®**) expressed as an intrabody **targeting aggregated TDP-43 (α-TDP-43 VecTab); (2) VecTabs®** expressed as a **secreted anti-OxPC scFv (α-OxPC VecTab), recognizing and neutralizing toxic OxPCs. Induced pluripotent stem cells**, derived from ALS patients, were **combined with microfluidics** for modelling ALS phenotypes as a **screening platform** for scFv efficacy. **Pathological ALS hallmarks** such as protein aggregation, impaired mitochondrial function and neurotoxicity were **evaluated in advanced *in-vitro* models (Fig. 2)**. Furthermore, we investigated the potential of scFv therapy in recovering gene expression changes in ALS, which are known to contribute to disease phenotypes.

These screening platforms facilitate the **personalized development of novel gene therapies** for **neurodegenerative diseases with translational potential to the clinic.**

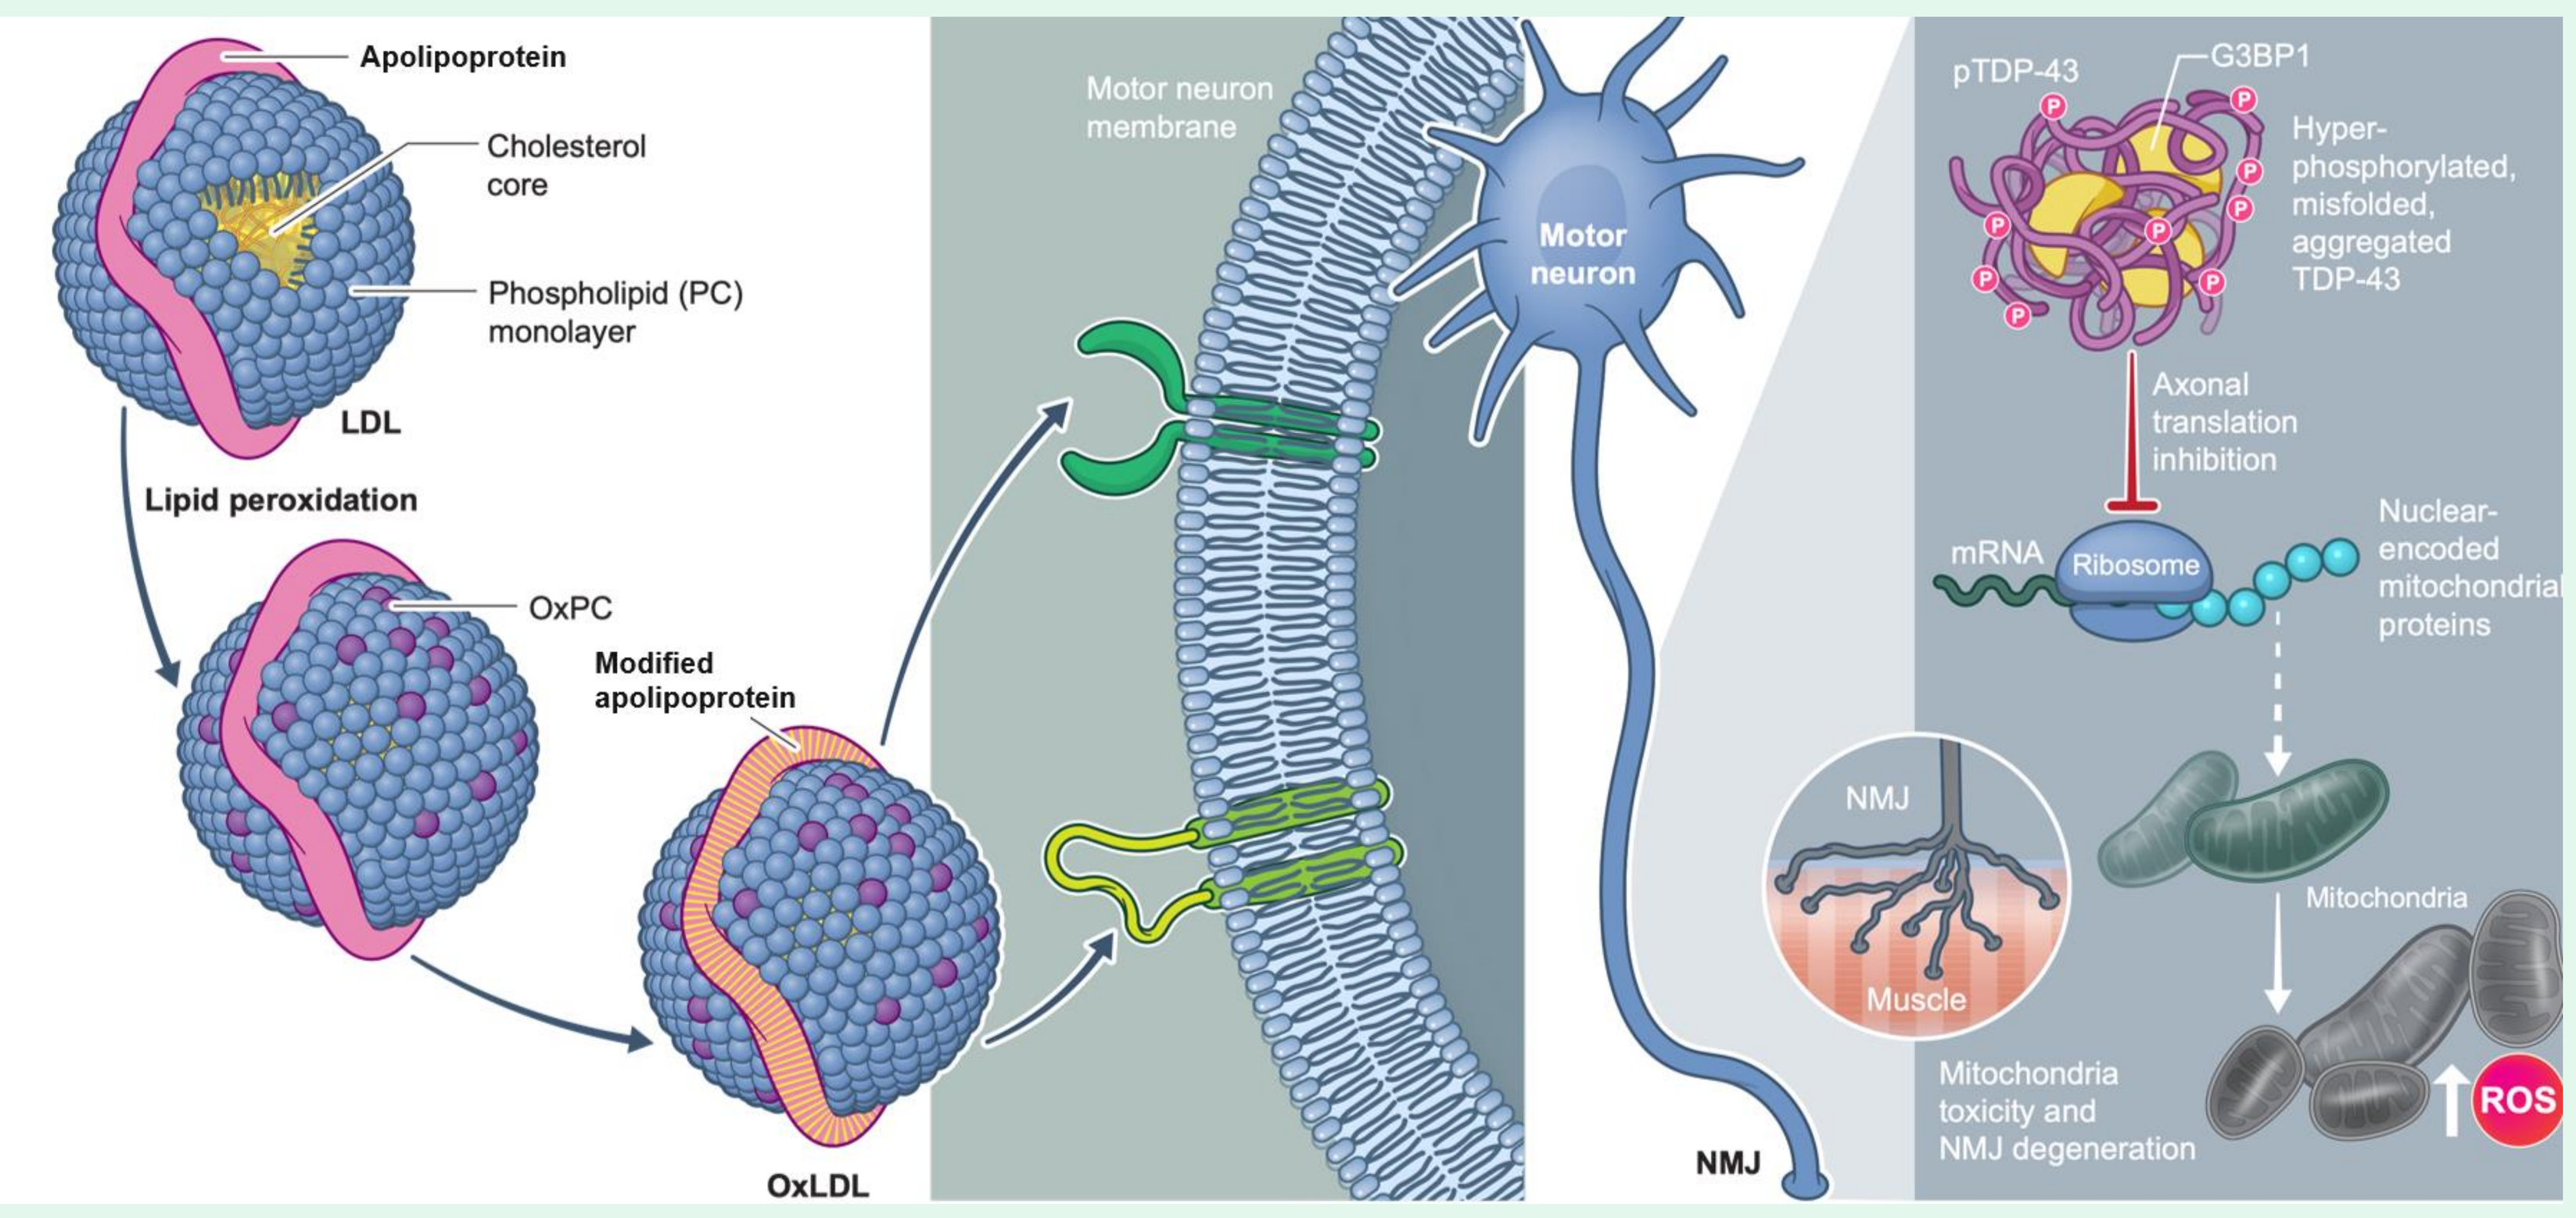
Apolipoprotein
Cholesterol core
Phospholipid (PC) monolayer
LDL
Lipid peroxidation
OxPC
Modified apolipoprotein
OxLDL
Motor neuron membrane
Motor neuron
NMJ
pTDP-43
G3BP1
Hyper-phosphorylated, misfolded, aggregated TDP-43
Axonal translation inhibition
mRNA
Ribosome
Nuclear-encoded mitochondrial proteins
NMJ
Muscle
Mitochondria
Mitochondria toxicity and NMJ degeneration
ROS

Figure 1: ALS neuronal pathology: OxPC formation and protein aggregation. In ALS, altered lipid metabolism and increased production of OxPC are present. These toxic species are transported on apolipoproteins, which become modified and can be recognized by receptors at the cellular membrane of motor neurons. OxPC are toxic to motor neurons, given their inability to efficiently metabolize fatty acids. As a consequence, these toxic species accumulate in the axonal segment of motor neurons, causing toxicity. This toxicity cascade increases the production of TDP-43 aggregates, which impair the normal translation of mitochondrial proteins and cause NMJ malfunctioning. Ultimately, oxidative stress alongside protein aggregation leads to the degeneration and neuronal death that characterizes ALS.

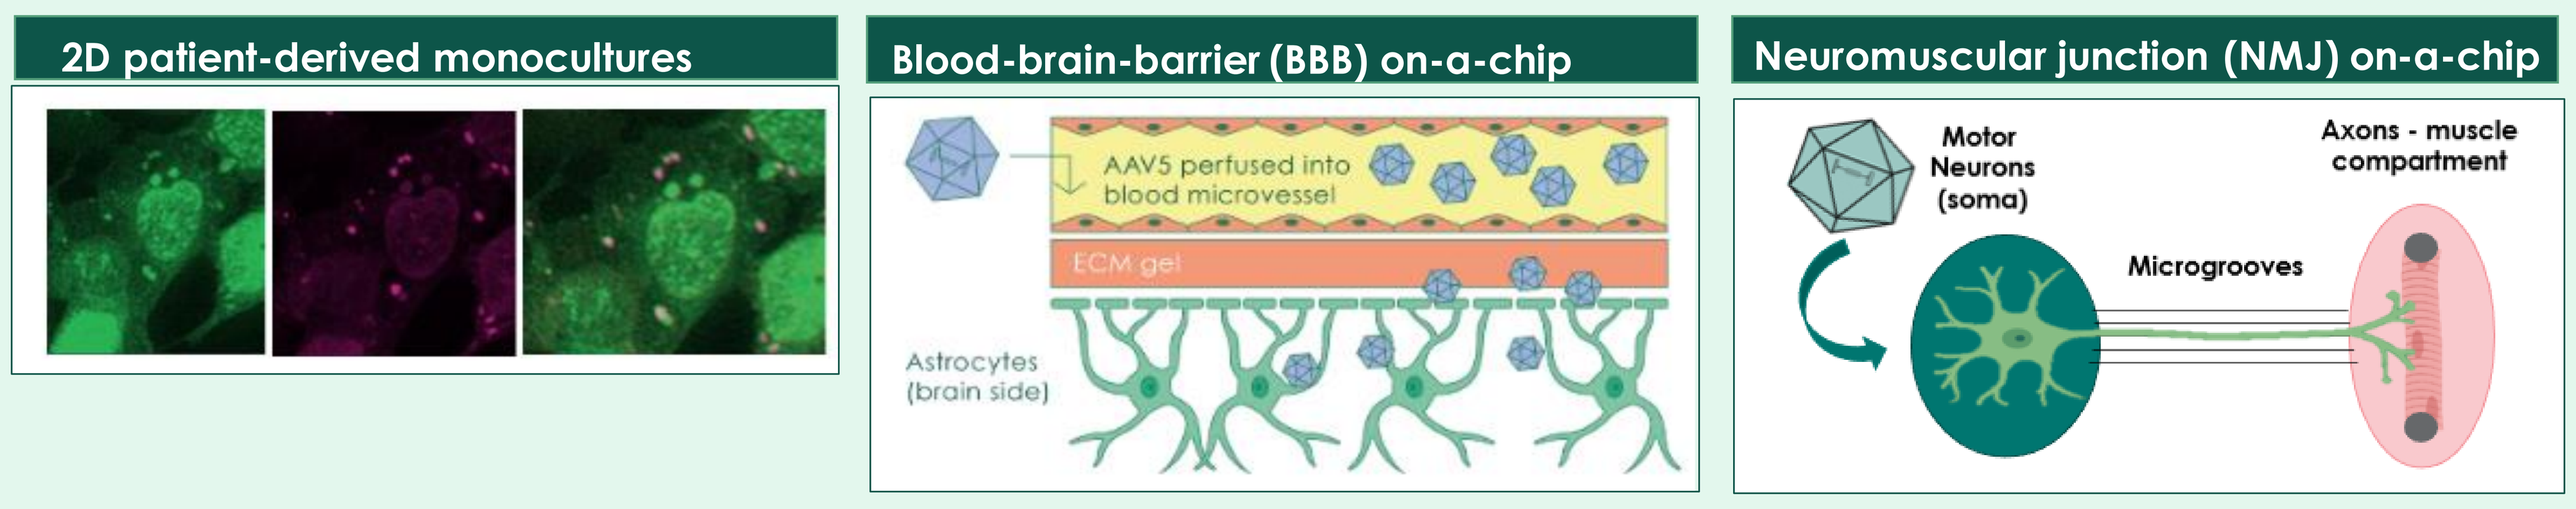
2D patient-derived monocultures
Blood-brain-barrier (BBB) on-a-chip
AAV5 perfused into blood microvessel
ECM gel
Astrocytes (brain side)
Neuromuscular junction (NMJ) on-a-chip
Motor Neurons (soma)
Axons - muscle compartment
Microgrooves

Figure 2: Advanced *in-vitro* models as a screening platform for VecTab efficacy.

Results

AAV-delivered VecTabs® are expressed in iPSC-derived neuronal cells and therapeutically relevant CNS areas

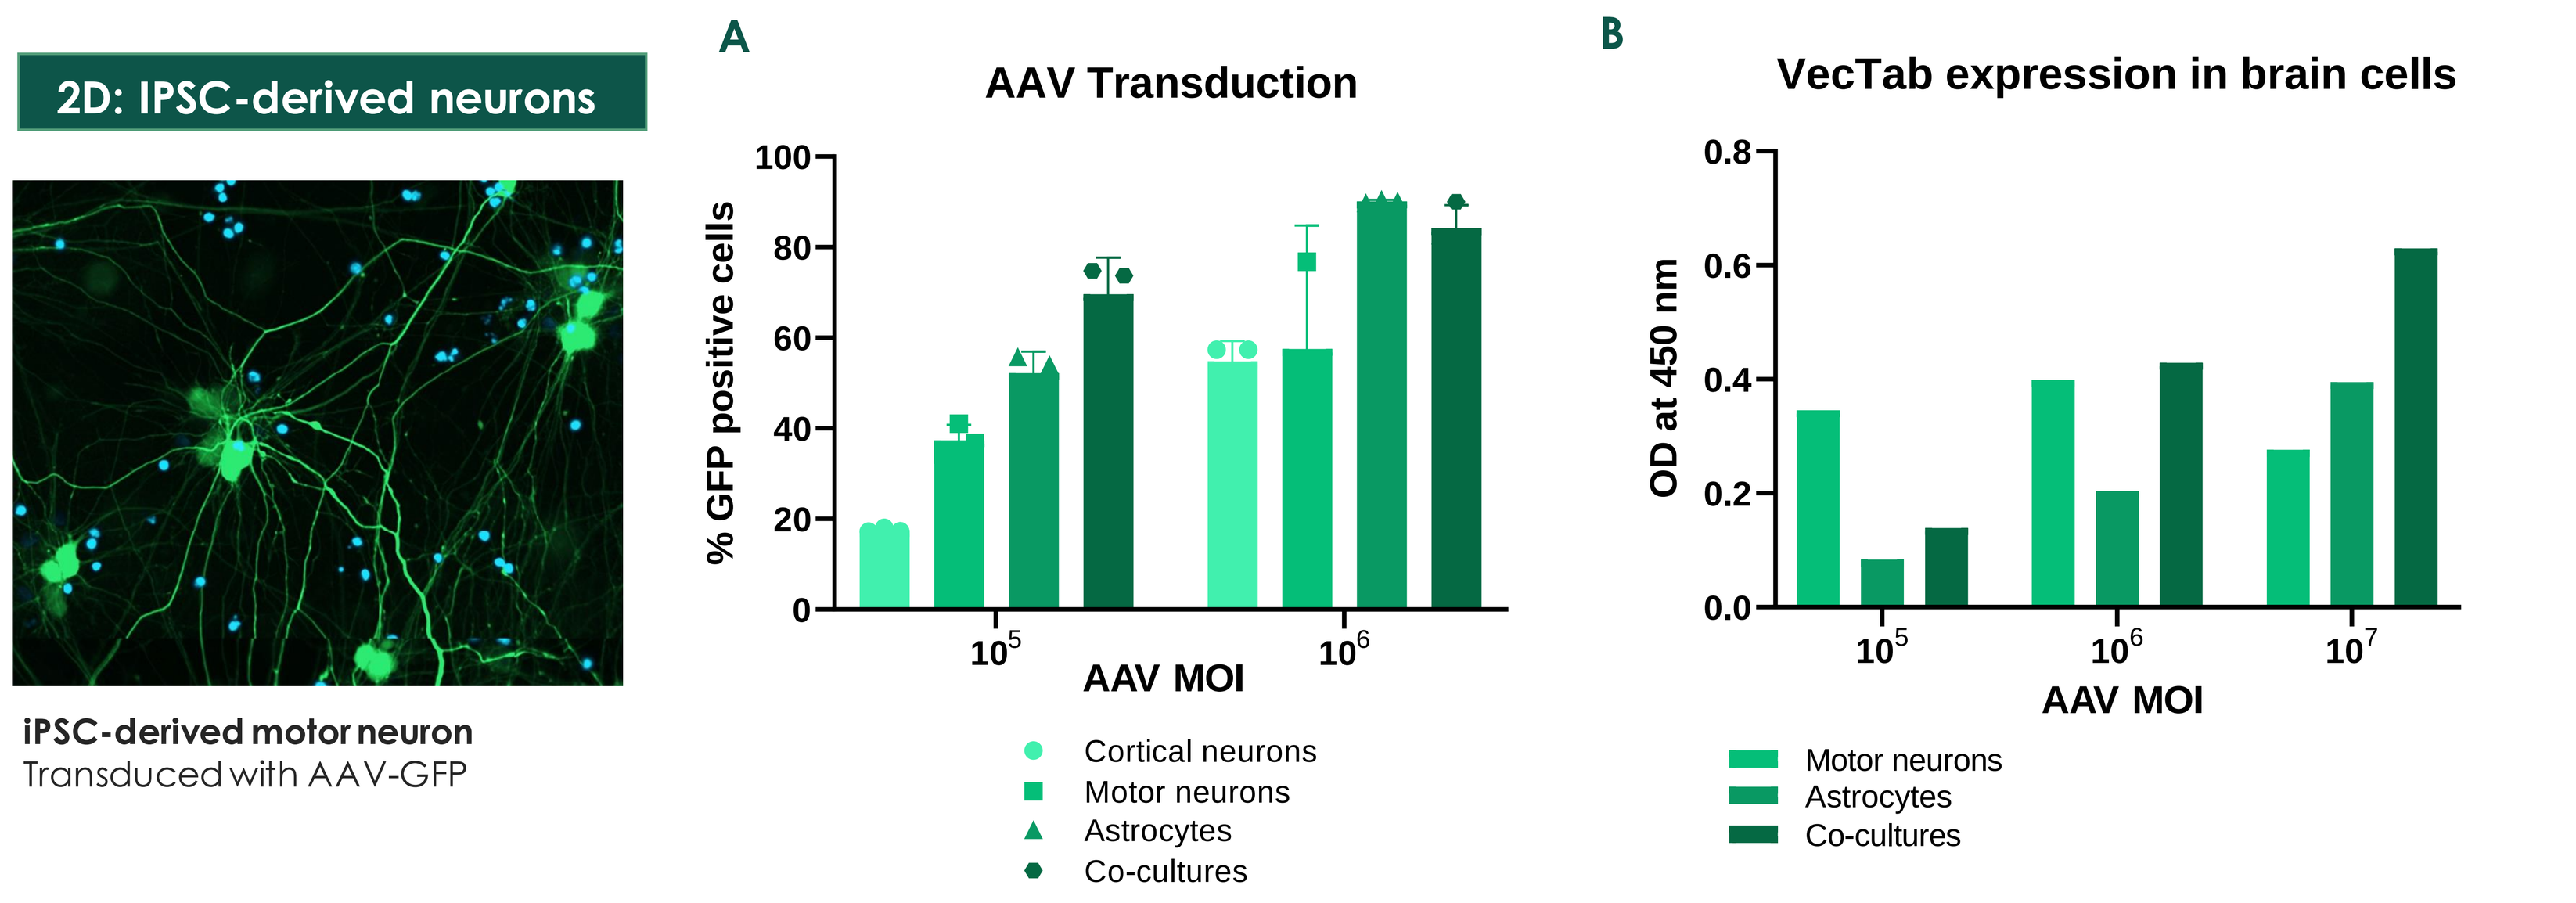
2D: IPSC-derived neurons
iPSC-derived motor neuron
Transduced with AAV-GFP
A
AAV Transduction
% GFP positive cells
AAV MOI
Cortical neurons
Motor neurons
Astrocytes
Co-cultures
B
VecTab expression in brain cells
OD at 450 nm
AAV MOI
Motor neurons
Astrocytes
Co-cultures

Figure 3: (A) Dose-dependent AAV transduction levels quantified by the percentage of GFP positive cells. **(B)** VecTabs® expression analysis by ELISA in disease affected cell types, showing a dose-dependent VecTabs® expression upon transduction.

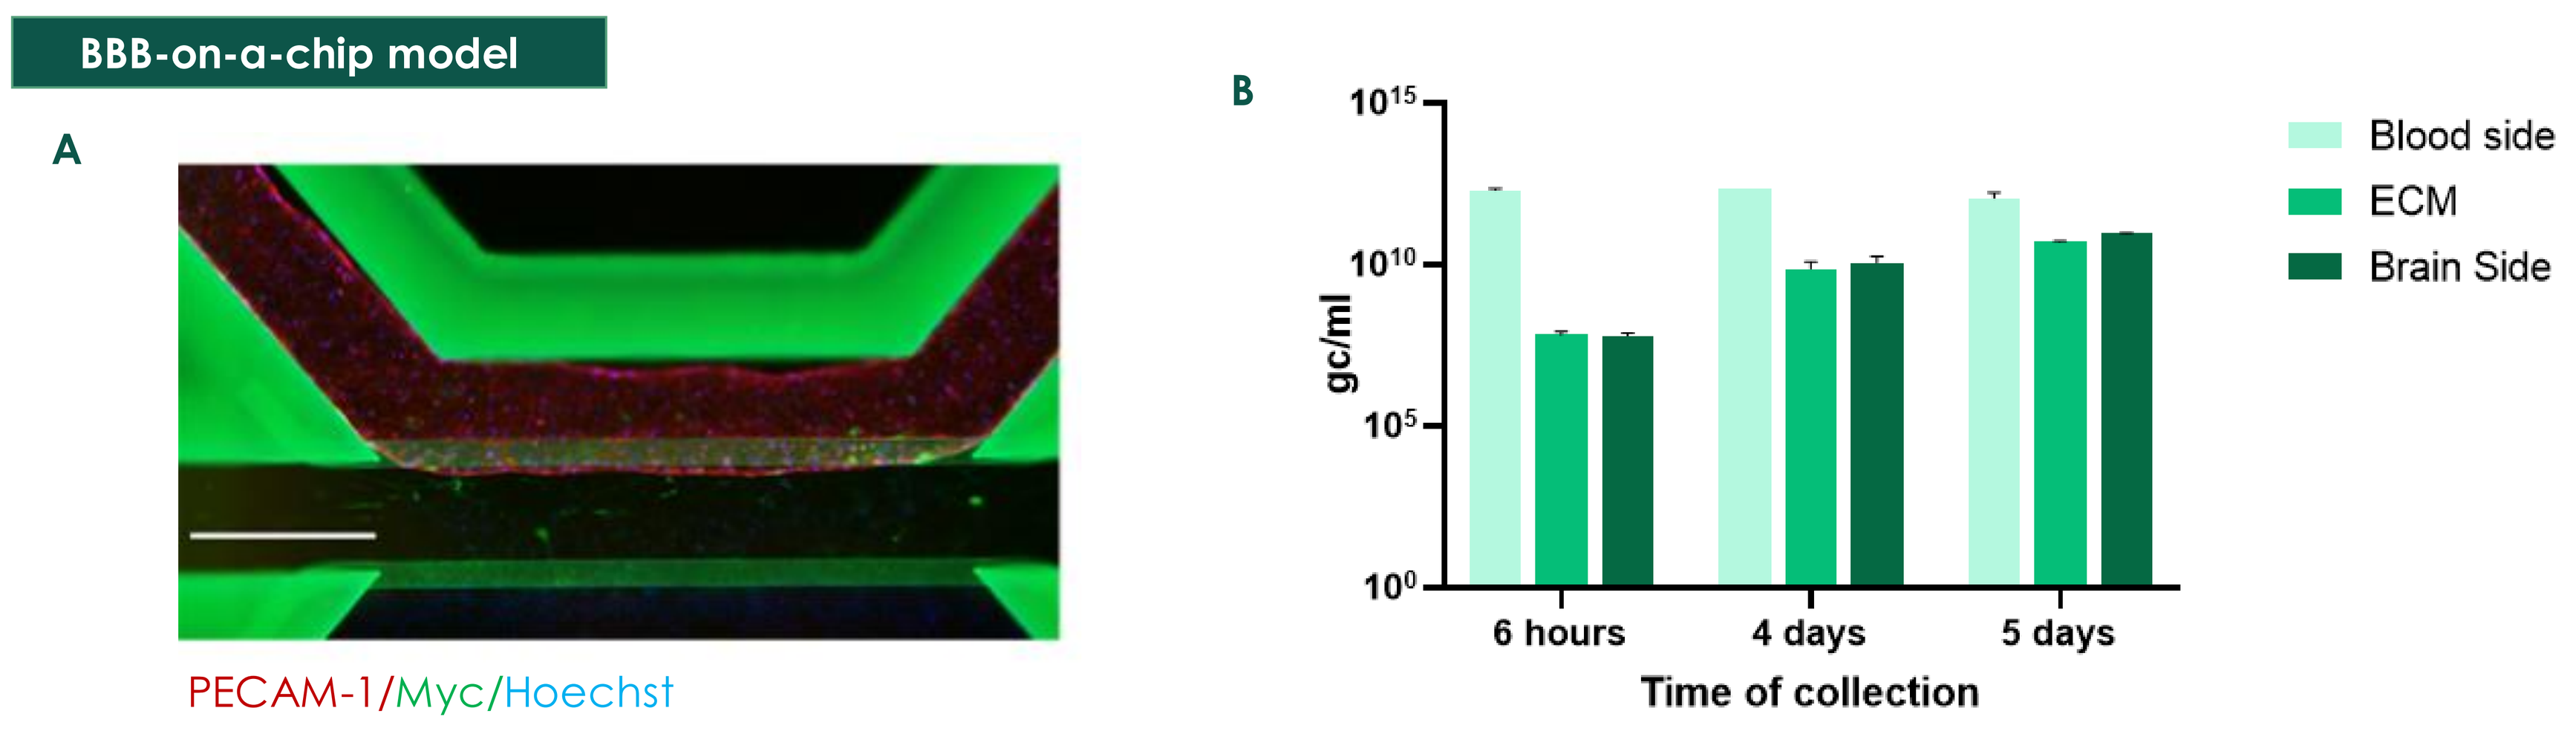
BBB-on-a-chip model
A
PECAM-1/Myc/Hoechst
B
gc/ml
Blood side
ECM
Brain Side
6 hours
4 days
5 days
Time of collection

Figure 4: (A) The BBB-model consists of human brain endothelial cells in the top channel (blood side) an ECM-gel and astrocytes on the bottom channel (brain side). PECAM-1 ((TRITC) was used as a marker of endothelial junction integrity; Myc (FITC) was used to detect AAV-VecTab expression and measure transduction efficiency (tag). **(B)** Samples from BBB-on-a-chip were collected at different timepoints post-transduction. RT-qPCR was performed to quantify AAV crossing from the blood to the brain side (ECM, extracellular matrix).

α-TDP-43 VecTab reverts ALS hallmarks in patient derived motor neurons

Gene expression

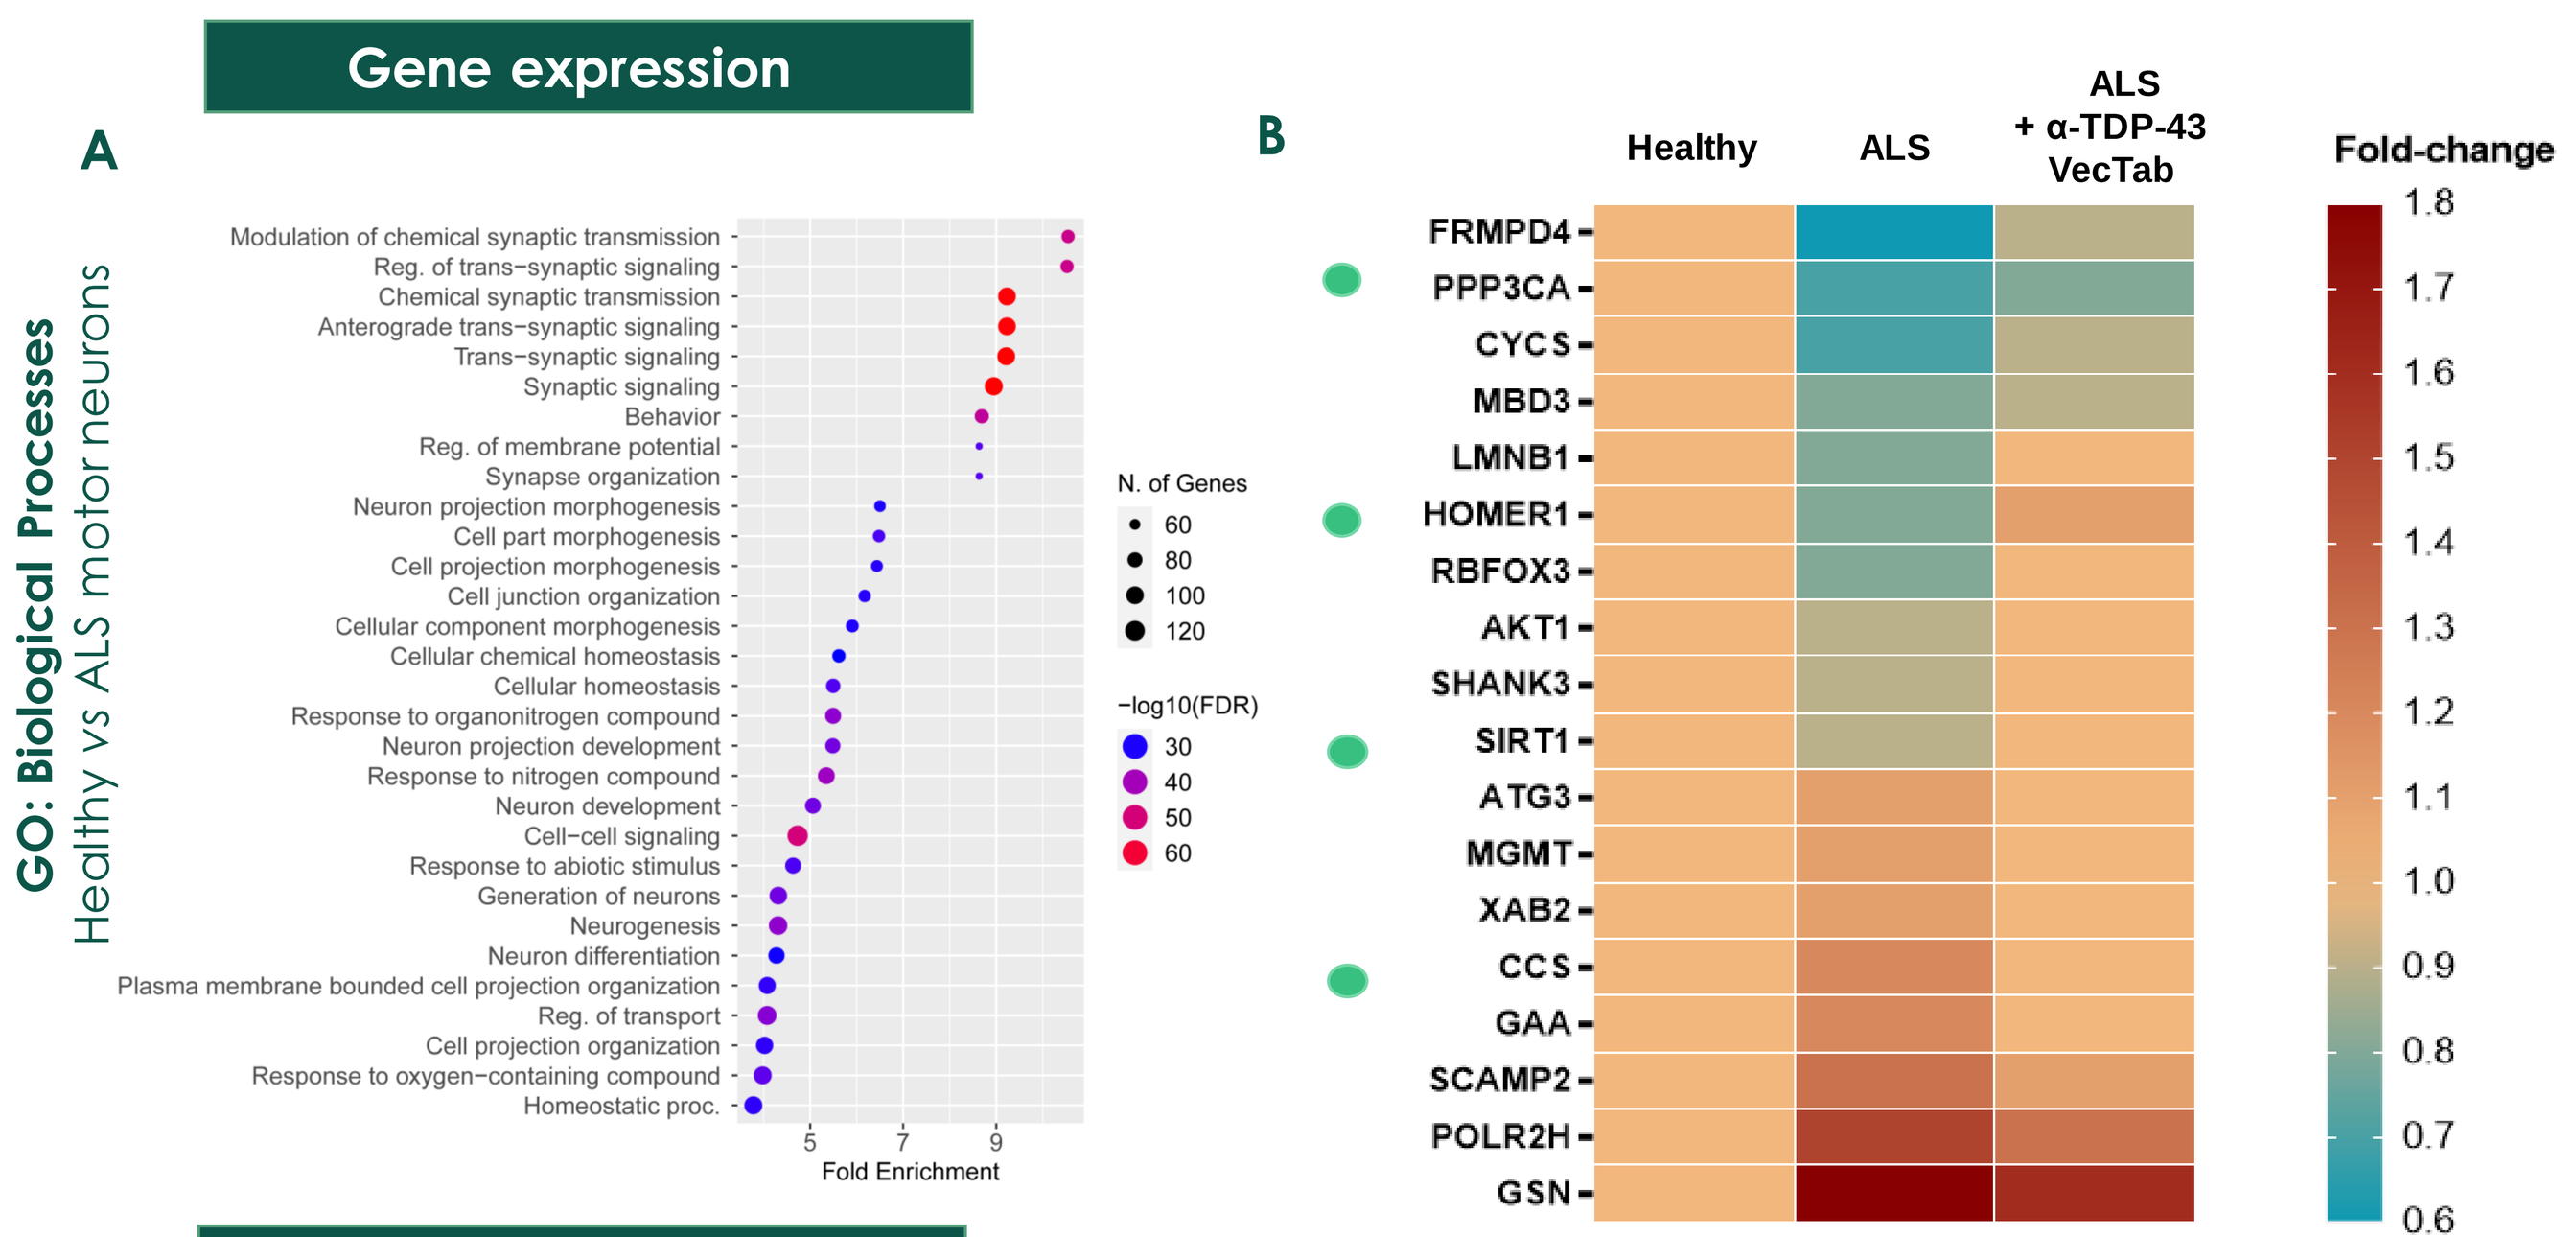
A
GO: Biological Processes
Healthy vs ALS motor neurons
Fold Enrichment
N. of Genes
-log10(FDR)
B
Healthy
ALS
ALS + α-TDP-43 VecTab
Fold-change
FRMPD4
PPP3CA
CYCS
MBD3
LMNB1
HOMER1
RBFOX3
AKT1
SHANK3
SIRT1
ATG3
MGMT
XAB2
CCS
GAA
SCAMP2
POLR2H
GSN

Figure 5: (A) Gene ontology (GO) analysis (GO category: Biological process) of differentially expressed genes in ALS-TDP-43 (M337V) motor neurons (Neuropathology panel, NanoString).
(B) Several transcripts, including direct targets/interactors of TDP-43 (marked in green) are recovered towards healthy levels after α-TDP-43 VecTab treatment. Cell values are normalized to healthy levels for visual representation.

Nuclear TDP-43 restoration

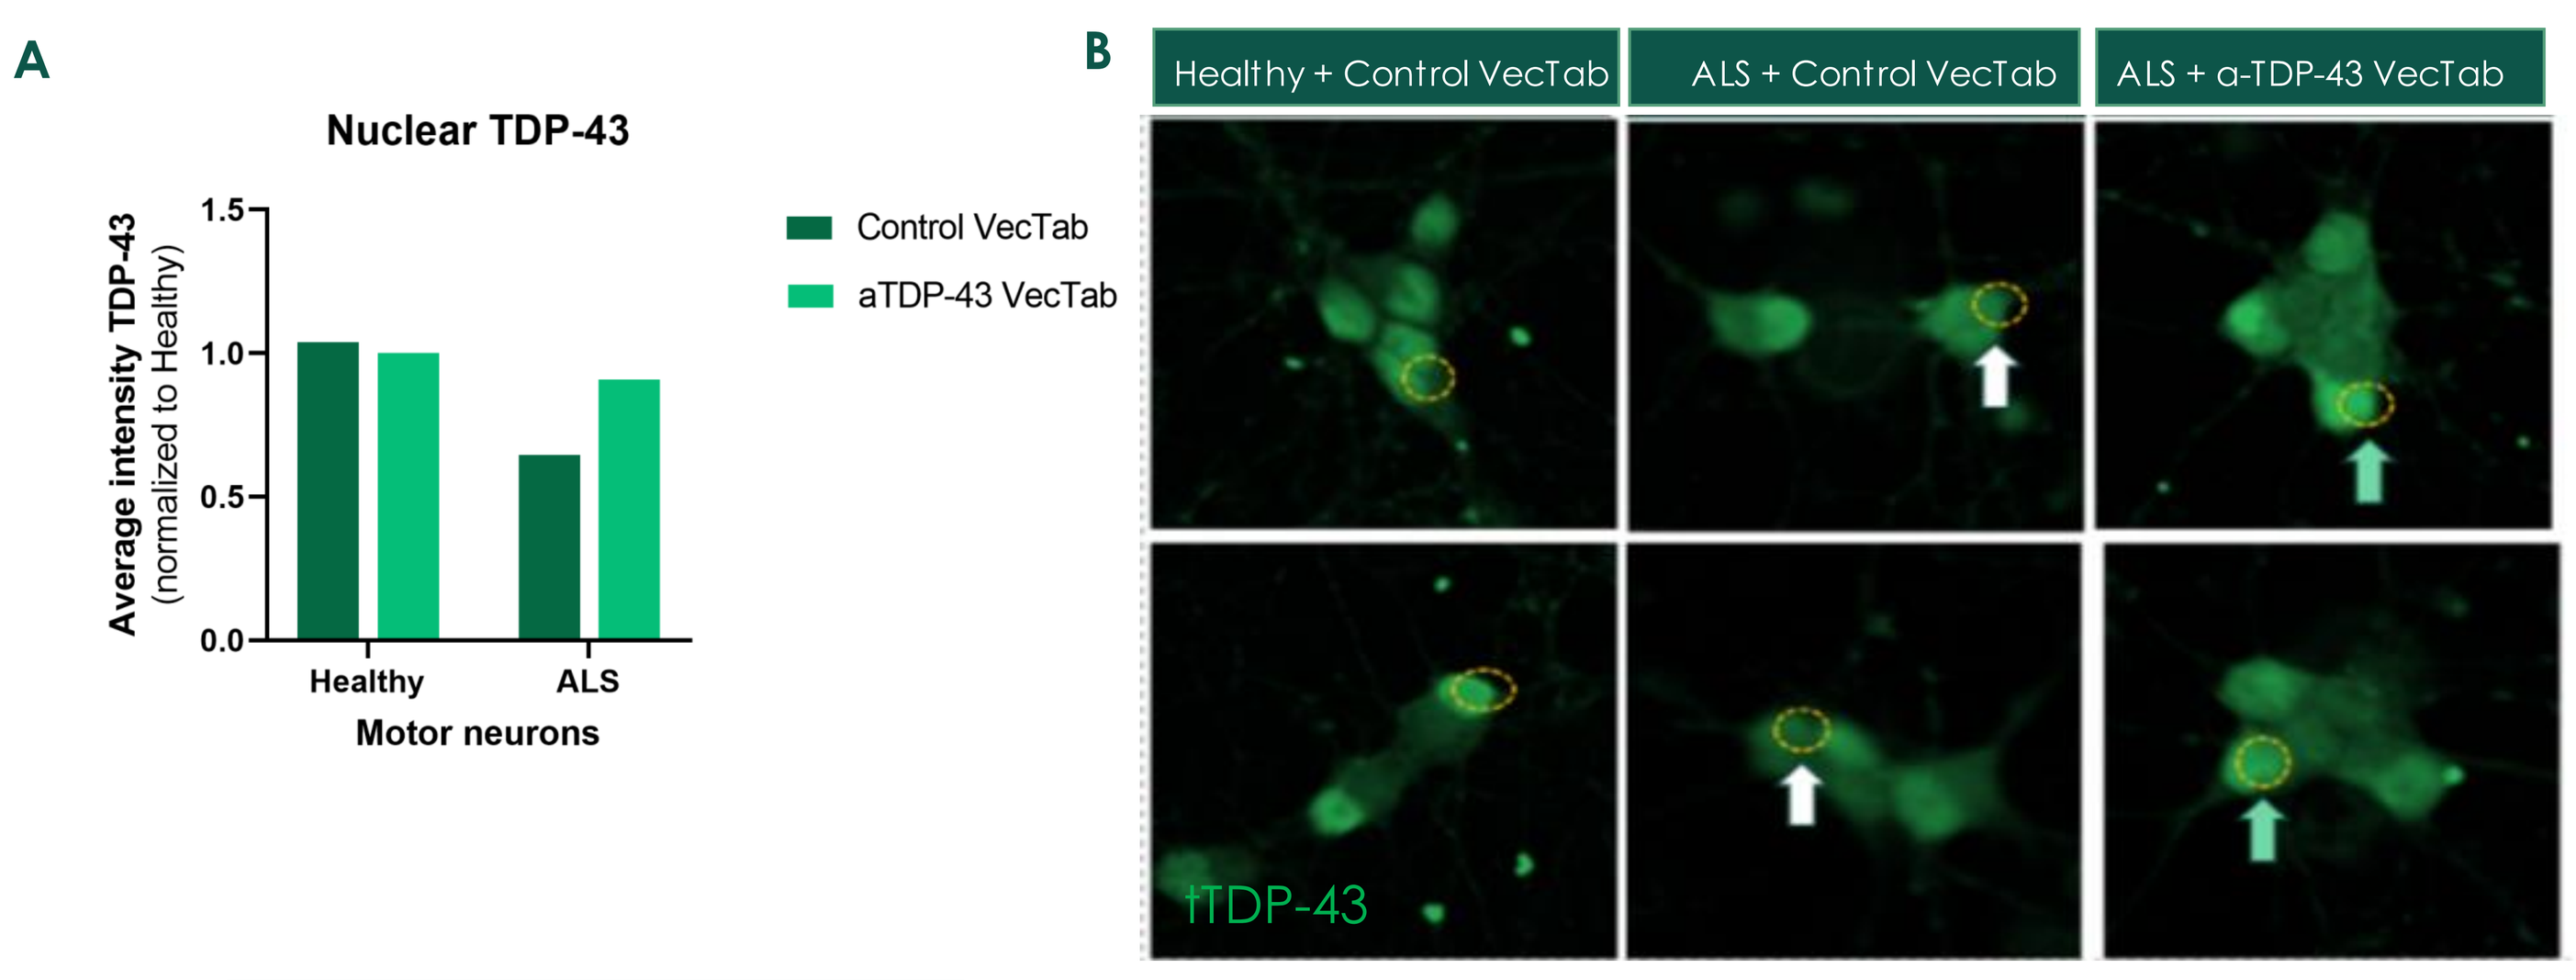
A
Nuclear TDP-43
Average intensity TDP-43 (normalized to Healthy)
Control VecTab
aTDP-43 VecTab
Healthy
ALS
Motor neurons
B
Healthy + Control VecTab
ALS + Control VecTab
ALS + a-TDP-43 VecTab
tTDP-43

Figure 6: (A) Average intensity of nuclear TDP43 of healthy and ALS –TDP-43 (M337V) motor neurons treated with α-TDP43-VecTab or control VecTab.
(B) Images representing expression of total TDP43 in motor neurons transduced with control or α-TDP-43 VecTab. Cell nuclei are stained with DAPI (blue), and TDP-43 in FITC (green). Arrows point nuclear total TDP-43 pattern (tTDP-43).

TDP-43 phosphorylation

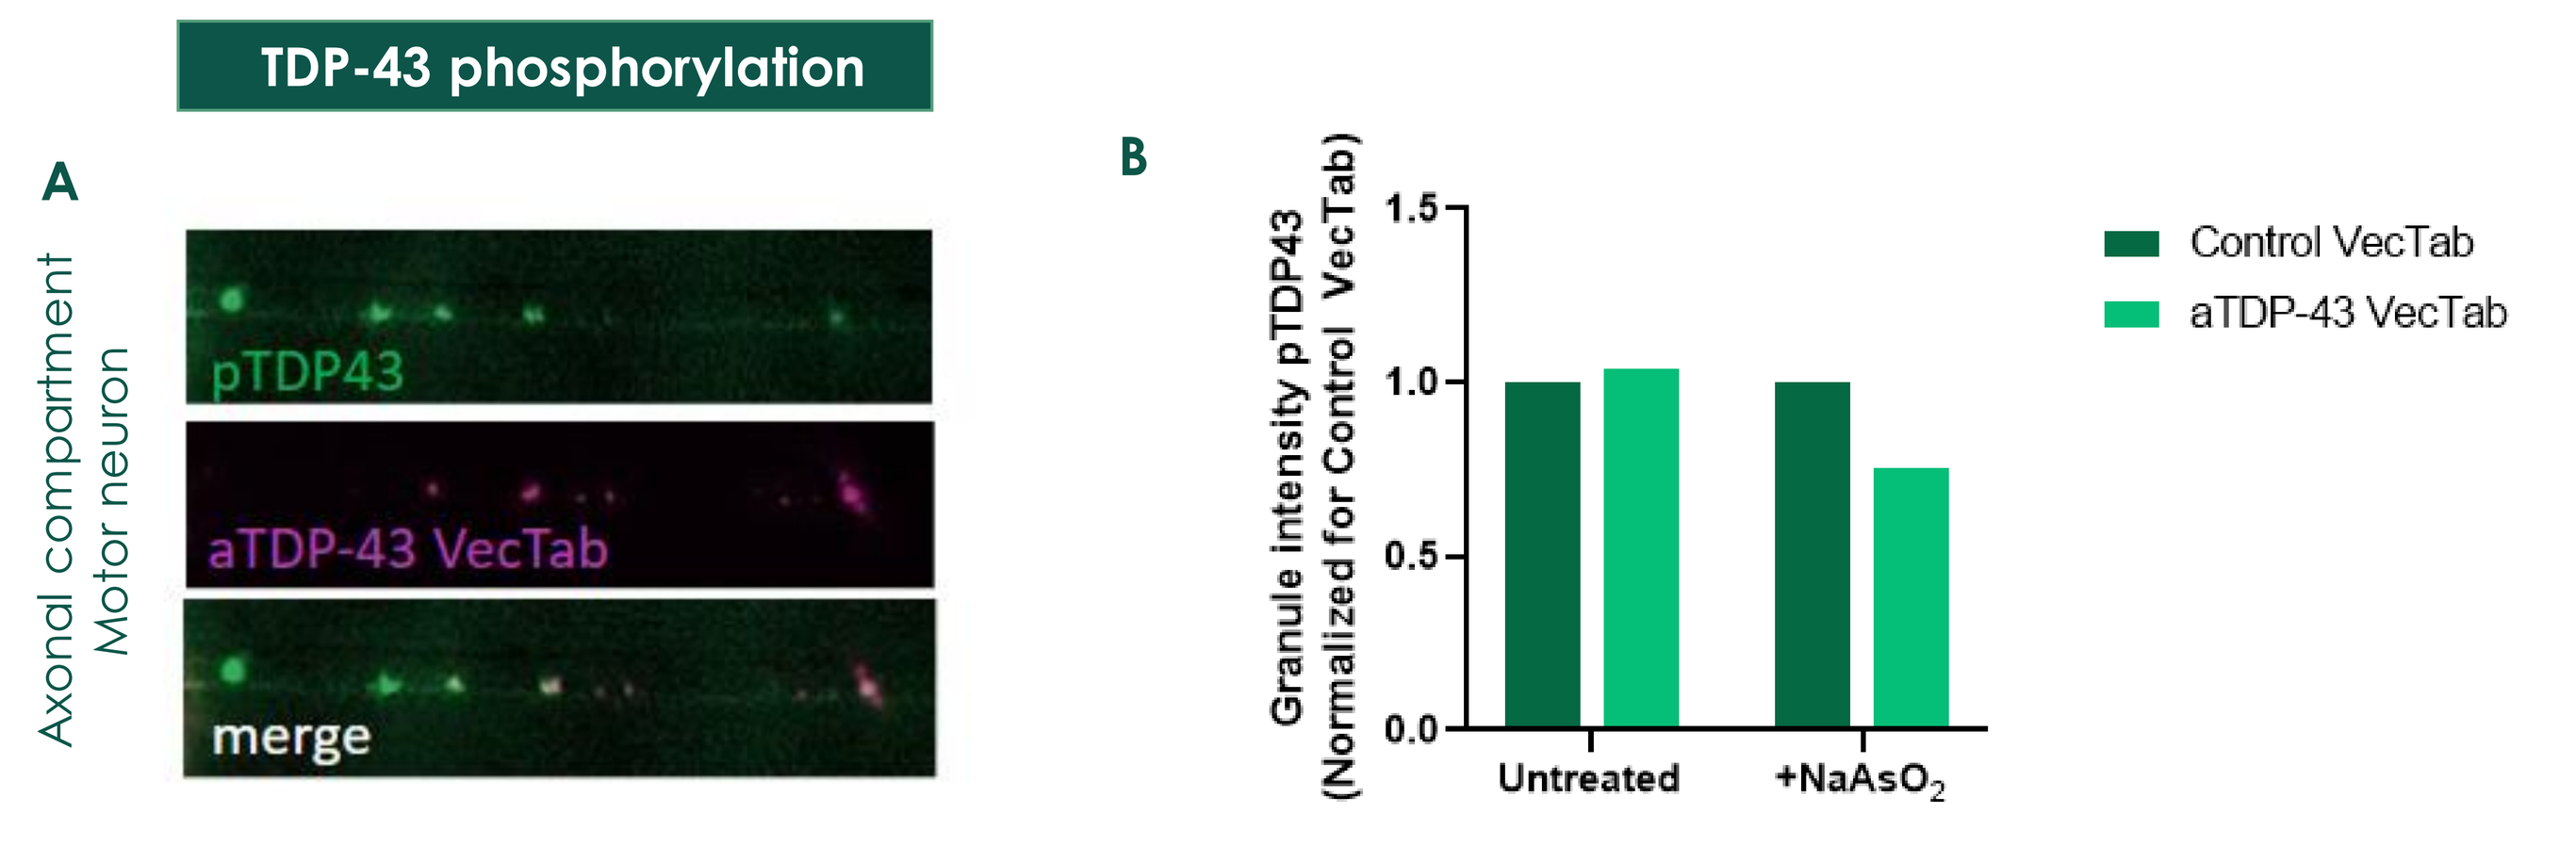
A
Axonal compartment
Motor neuron
pTDP43
aTDP-43 VecTab
merge
B
Granule intensity pTDP43 (Normalized for Control VecTab)
Control VecTab
aTDP-43 VecTab
Untreated
+NaAsO2

Figure 7: (A) pTDP-43 was detected in the axons of the mutant motor neurons (FITC), where it shows co-localization with α-TDP-43 VecTab (Cy5) (merge). **(B)** pTDP-43 profile in healthy motor neurons treated with control or α-TDP-43 VecTab, under physiological and stress conditions (NaAsO2, sodium arsenite).

α-OxPC VecTab reverts OxPC-toxicity in patient derived motor neurons

Gene expression

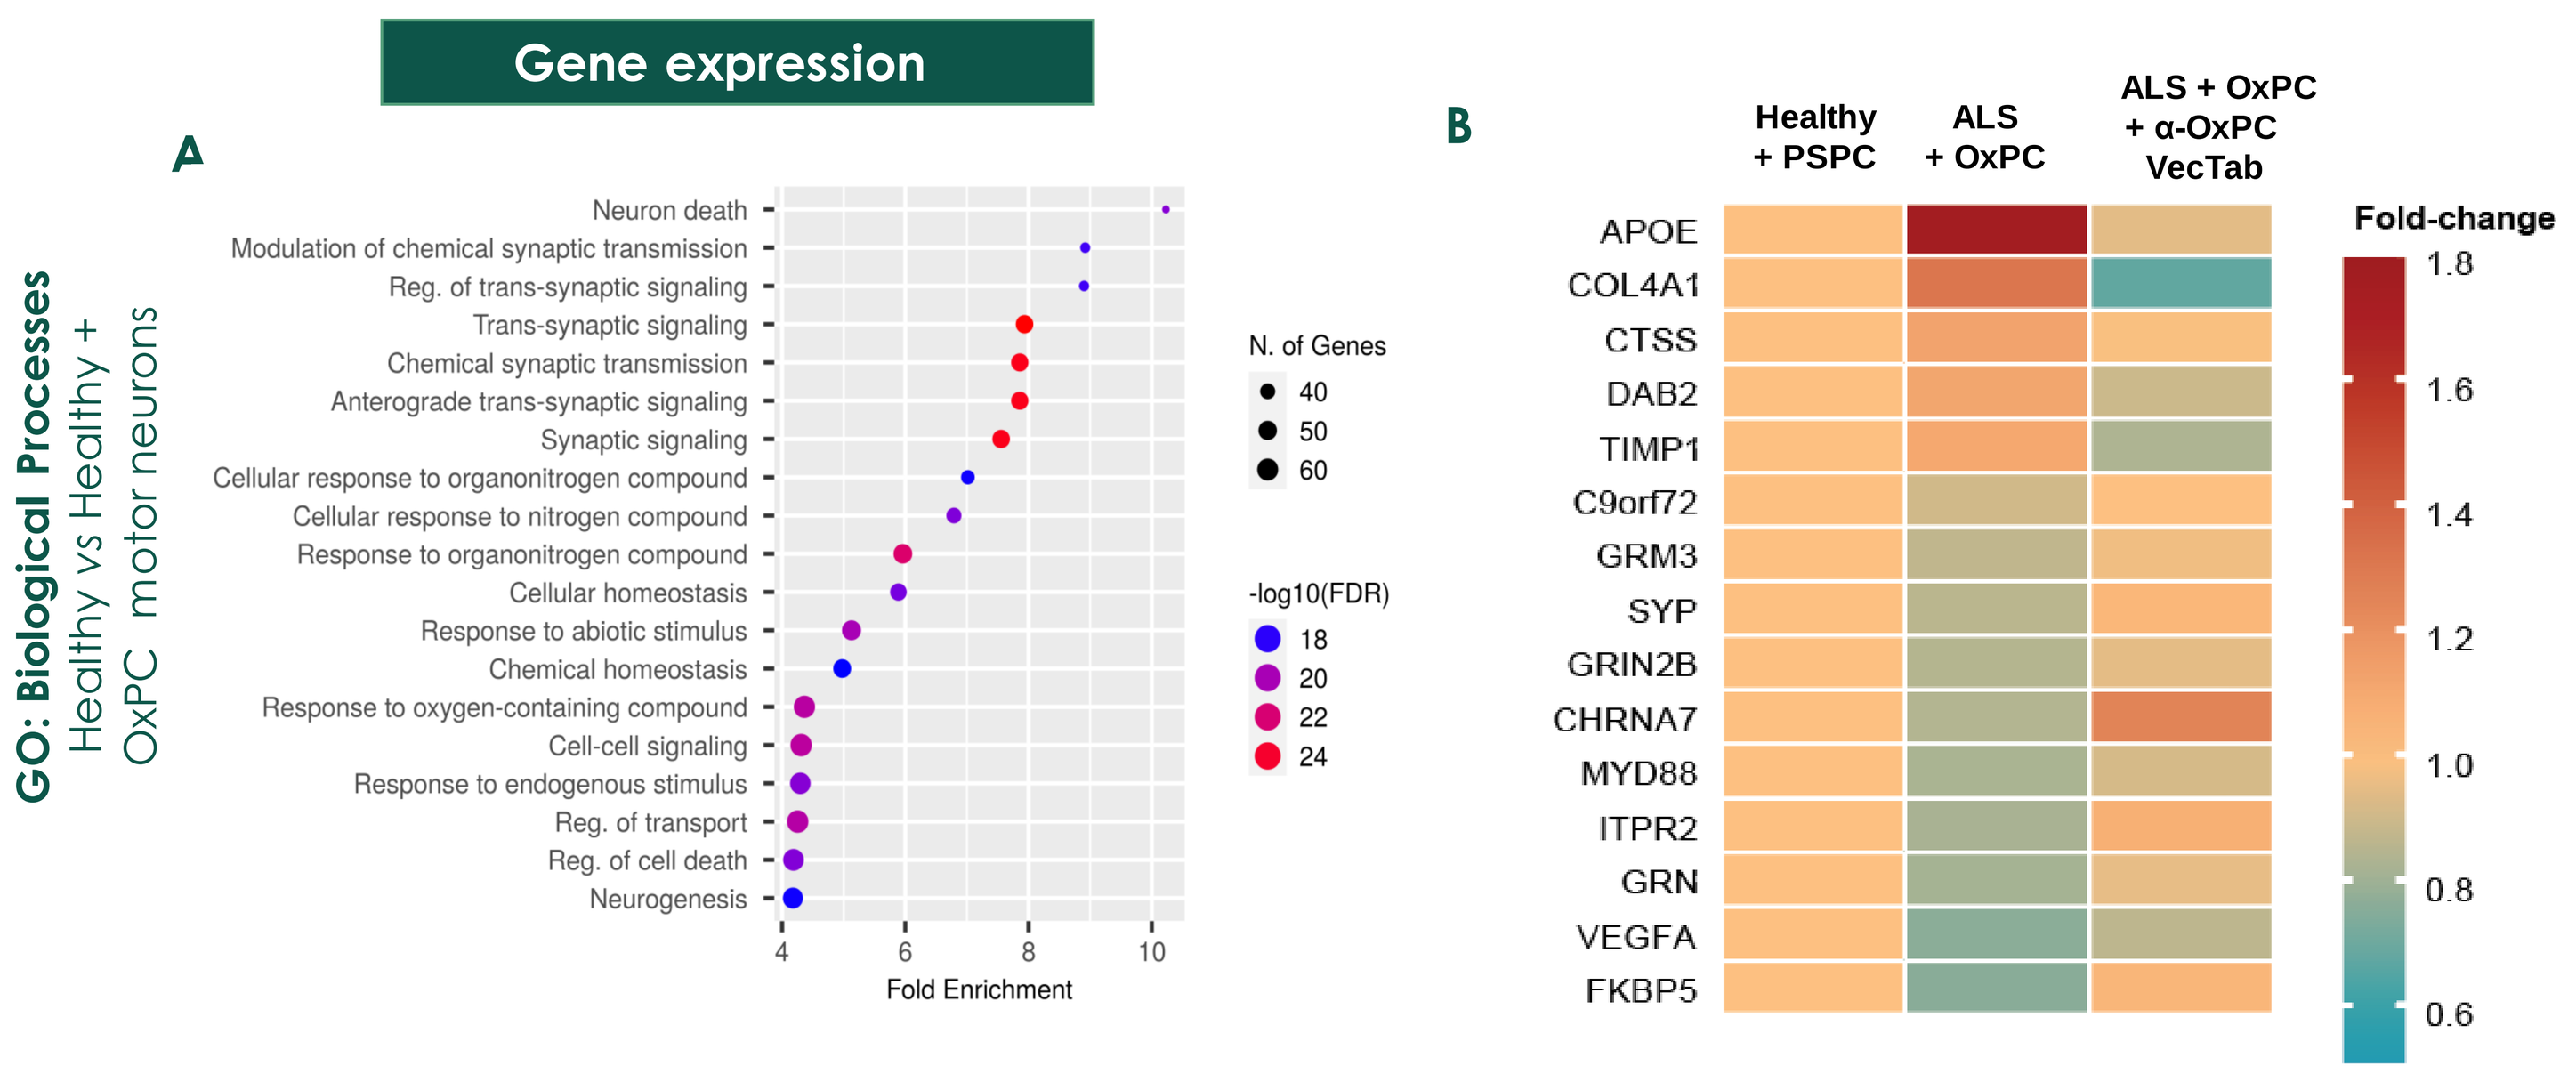
A
GO: Biological Processes
Healthy vs Healthy + OxPC motor neurons
Fold Enrichment
N. of Genes
-log10(FDR)
B
Healthy + PSPC
ALS + OxPC
ALS + OxPC + α-OxPC VecTab
Fold-change
APOE
COL4A1
CTSS
DAB2
TIMP1
C9orf72
GRM3
SYP
GRIN2B
CHRNA7
MYD88
ITPR2
GRN
VEGFA
FKBP5

Figure 8: (A) Gene ontology (GO) analysis (GO category: Biological process) of differentially expressed genes in ALS-SOD1 (G93A) motor neurons (Neuropathology panel, NanoString).
(B) Gene expression is deregulated following OxPC treatment of healthy motor neurons and can be normalized with α-OxPC VecTab. Cell values are normalized to healthy PSPC condition for visual representation.

Neuronal outgrowth

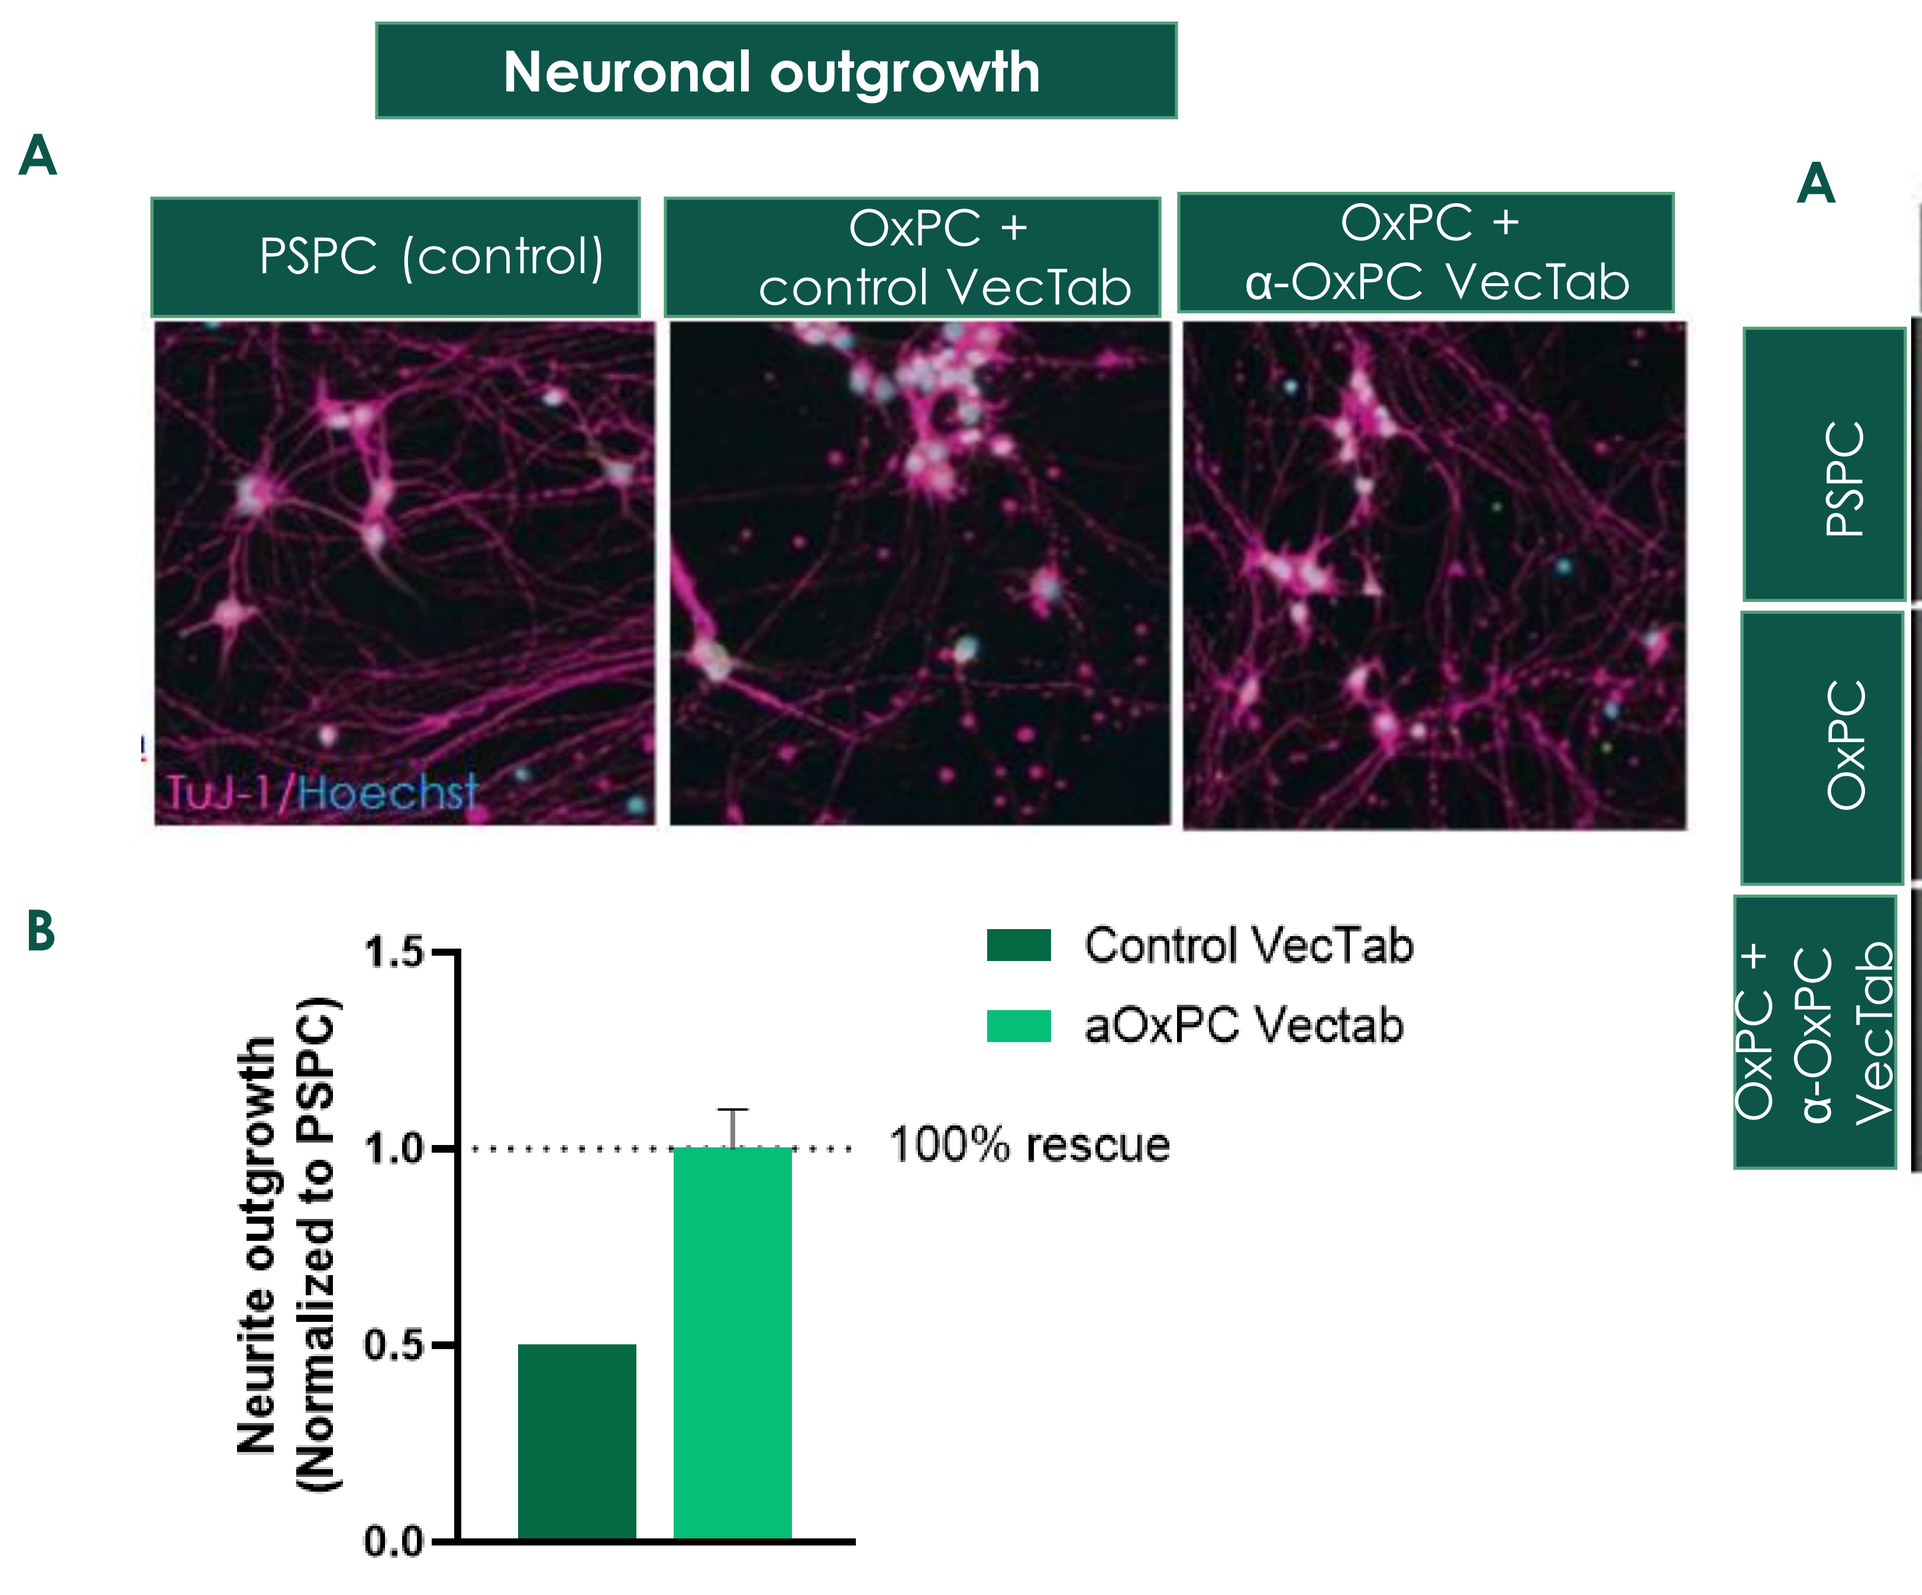
A
PSPC (control)
OxPC + control VecTab
OxPC + α-OxPC VecTab
TuJ-1/Hoechst
B
Neurite outgrowth (Normalized to PSPC)
Control VecTab
aOxPC Vectab
100% rescue

Figure 9: (A) Representative images of OxPC-induced neuronal toxicity and α-OxPC VecTab rescue in healthy motor neurons. Tuj-1 (Cy5) was used as a marker for neurite outgrowth. **(B)** Bar graph showing the rescue effect of α-OxPC VecTab on OxPC-induced toxicity in healthy motor neurons. Values are shown in reference to PSPC condition (control).

Distal neuronal pathology

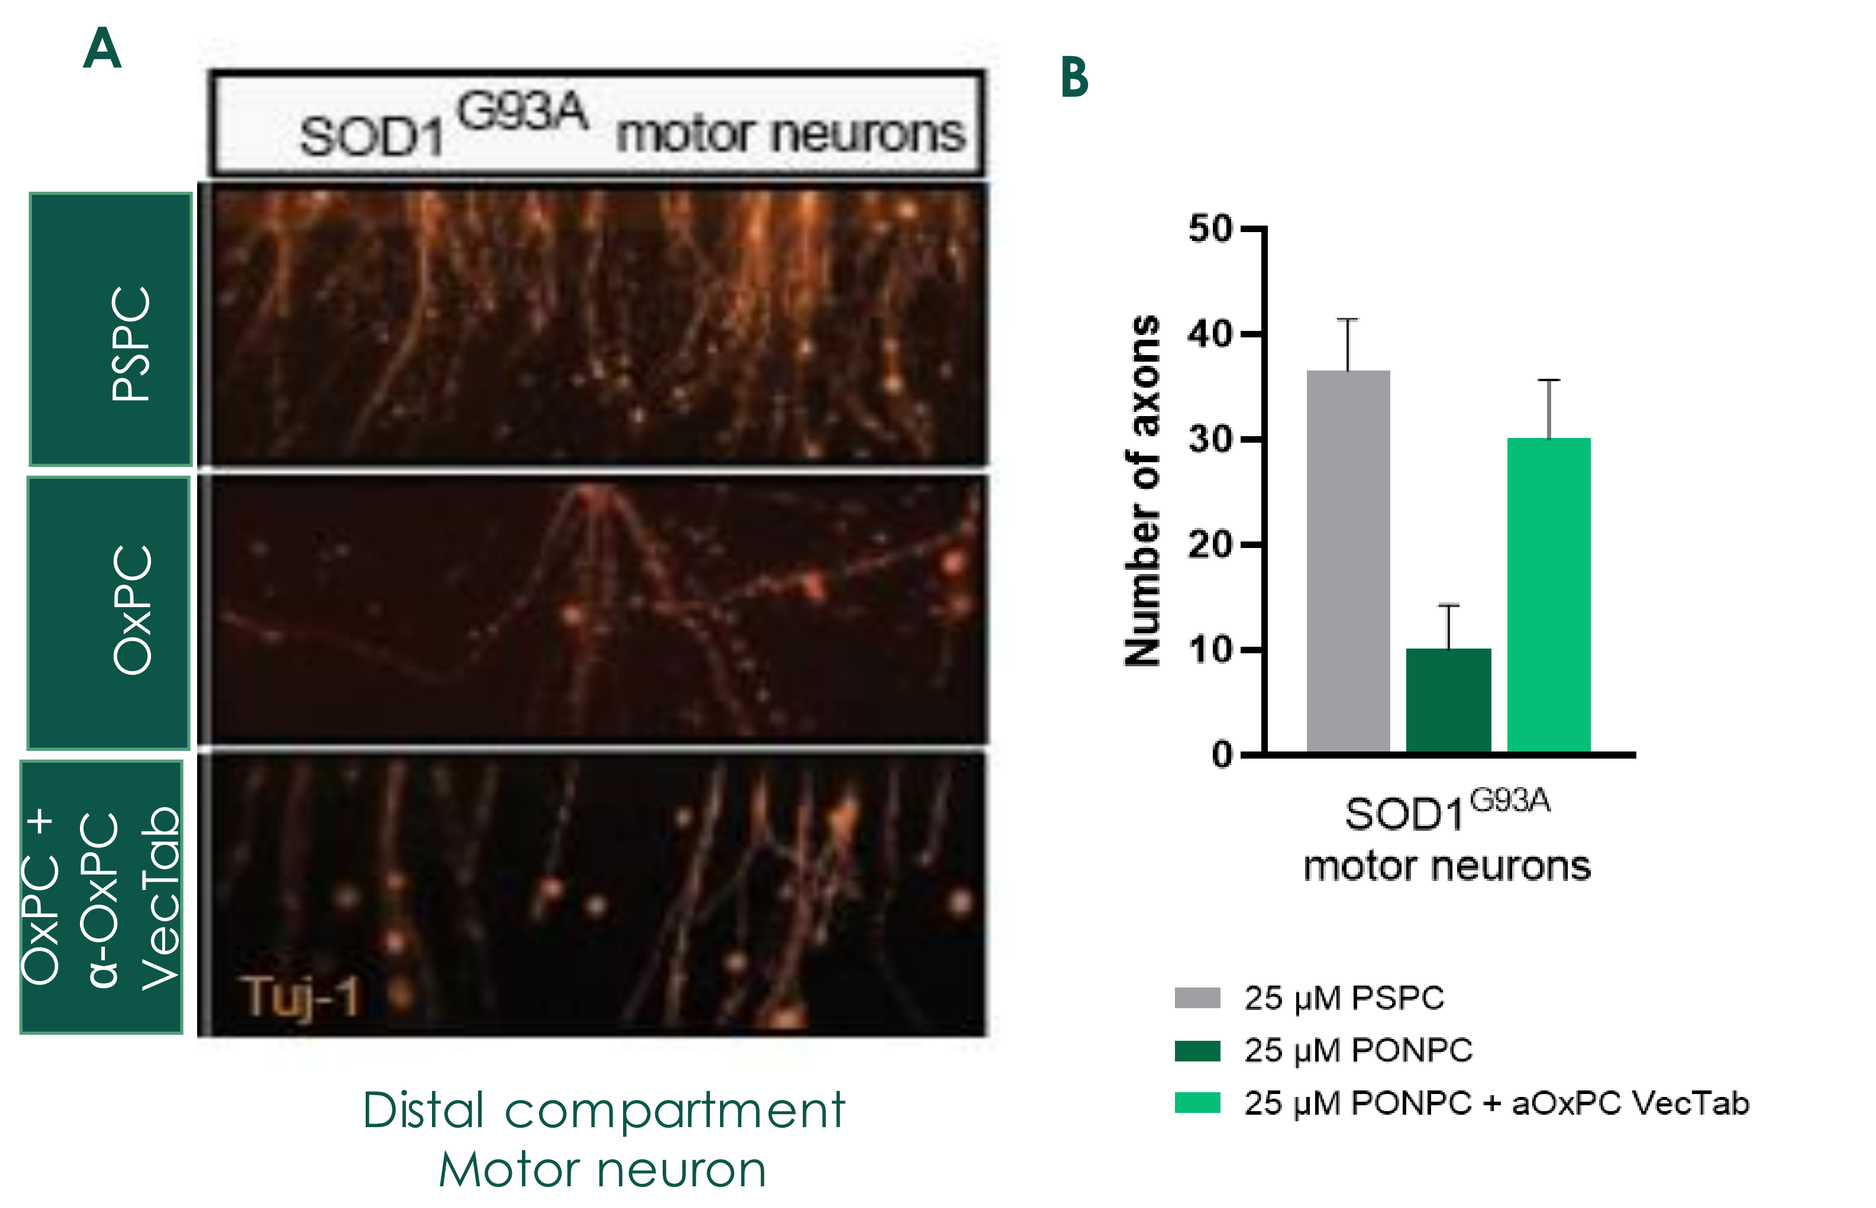
A
SOD1 G93A motor neurons
PSPC
OxPC
OxPC + α-OxPC VecTab
Tuj-1
Distal compartment
Motor neuron
B
Number of axons
SOD1 G93A motor neurons
25 µM PSPC
25 µM PONPC
25 µM PONPC + aOxPC VecTab

Figure 10: (A) Representative images of OxPC-induced neuronal toxicity and α-OxPC VecTab rescue in SOD1 (G93A) motor neurons. Tuj-1 (TRITC) was used as a marker for neurite outgrowth. **(B)** OxPC-induced distal neuronal toxicity is recovered with α-OxPC VecTab in ALS-SOD1 (G93A) motor neurons. Bars represent the mean and values are normalized to PSPC levels. The number of axons was used as a parameter to evaluate neuronal toxicity.

Summary and Future steps

- ❑ Successful AAV transduction and VecTabs® expression were demonstrated in iPSC-derived models, including different CNS regions.
- ❑ VecTabs® successfully reverted gene expression changes, pathological TDP-43 aggregates and OxPC-mediated neuronal toxicity in different *in-vitro* ALS systems.
- ❑ Our models provide a framework for pre-clinical screening of different therapeutic strategies (incl. VecTabs®) targeting CNS diseases.

VECTORY

33rd international symposium on ALS/MND